gBoostTM, a cell-selective gene expression enhancer

Mak Inoue, PhD, Infirmacea Inc. (Chiba, Japan)

Introduction

Gene therapy is a ground-breaking therapeutic approach. Most of gene therapies rely on viral vectors, especially AAVs (adeno-associated virus), which come with notable issues, including safety concerns and extremely high therapeutic costs, and therefore limited applicability - mainly restricted to rare genetic disorders. For its wide therapeutic application, these drawbacks should be overcome. Infirmacea has developed gBoostTM, a cell-selective gene expression enhancer as a revolutionary excipients, that makes gene therapy safer and more affordable. gBoostTM enhances gene expression regardless of the delivery system, either non-viral or viral. Contrary to conventional approach, which focused on cellular targeting/uptake, gBoostTM exhibit its effects on cellular protective responses that are triggered by transferred genetic materials.

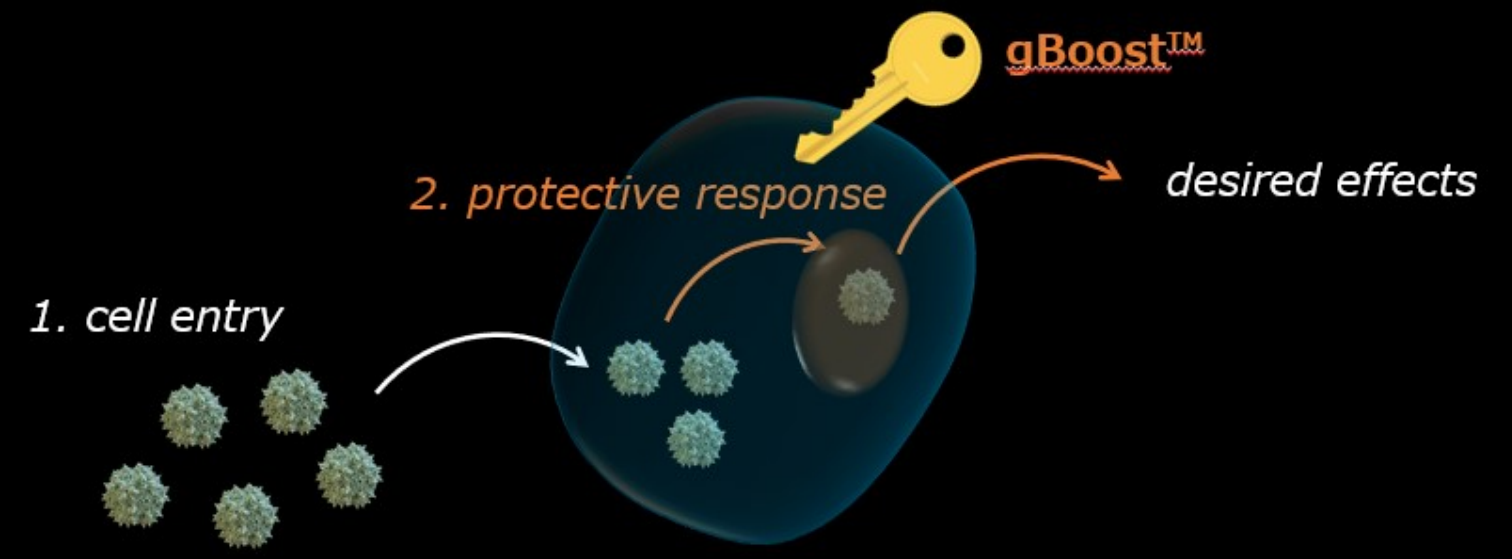
gBoost™
2. protective response
desired effects
1. cell entry

Effects of gBoostTM on viral gene expression

Methods: Human subcutaneous fat tissue (~70mg, obtained as a surgical waste) was injected with 5uL of AAV_EGFP (2 x 10^9vg) containing gBoost (or without gBoost as control). The tissue was cultured in a device (WO2024162464) for the indicated period, and subjected to microscopy analysis (Keyence, BZ-9000, FITC filter, focus x10)

AAV8_EGFP_6w after gene transfer

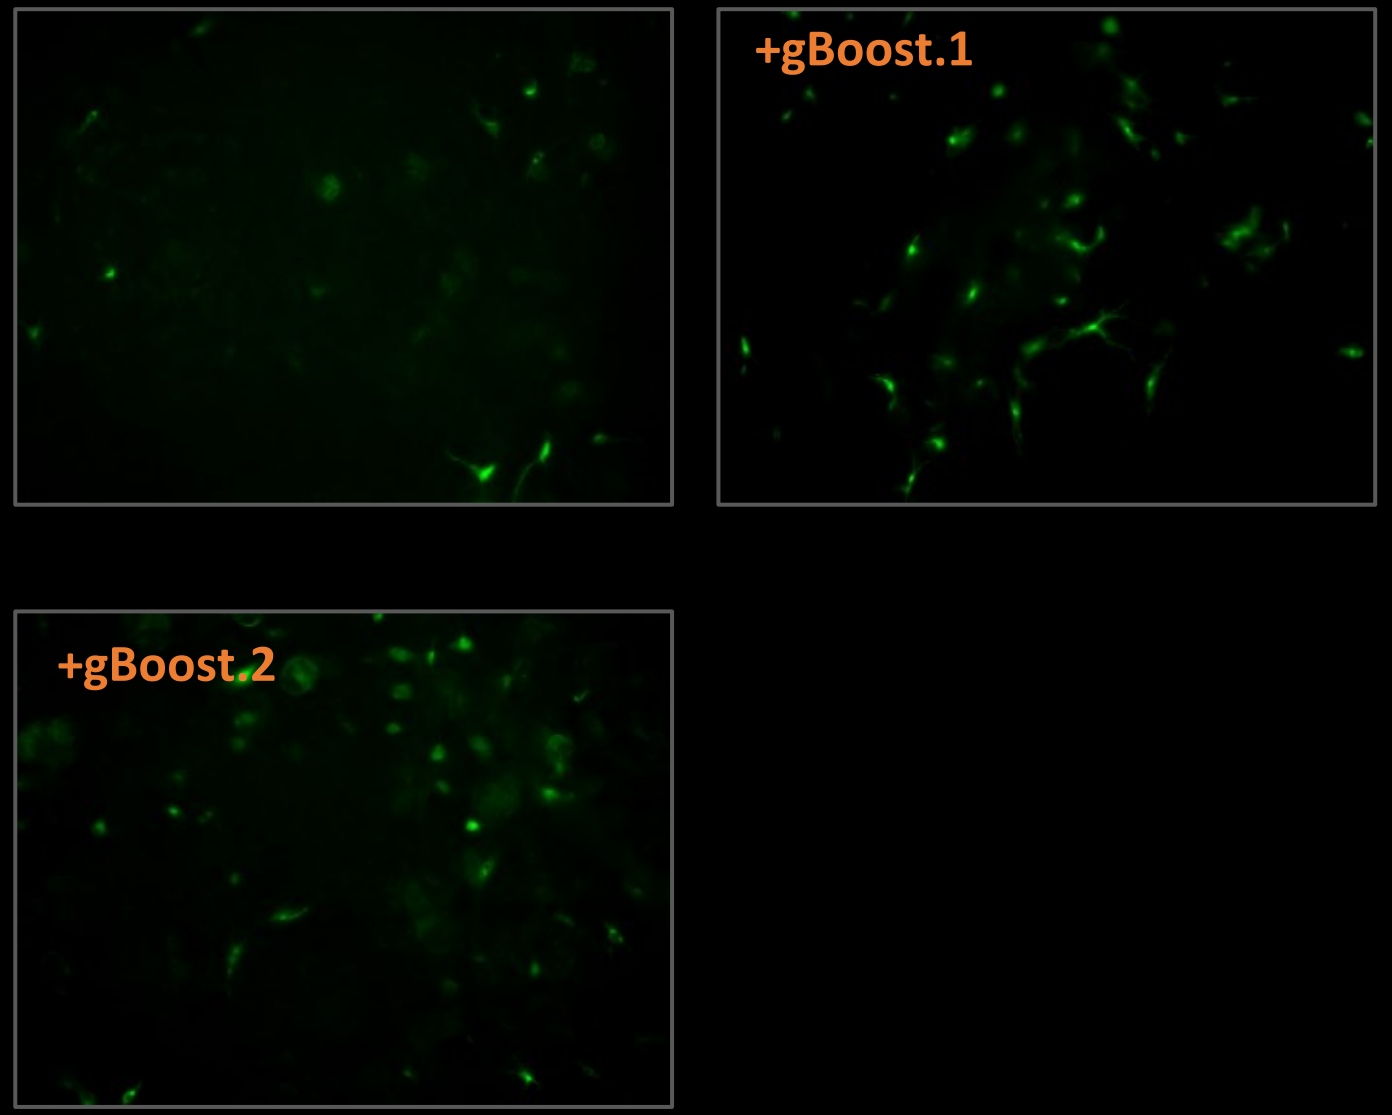
+gBoost.1
+gBoost.2

AAV5_EGFP_6w after gene transfer

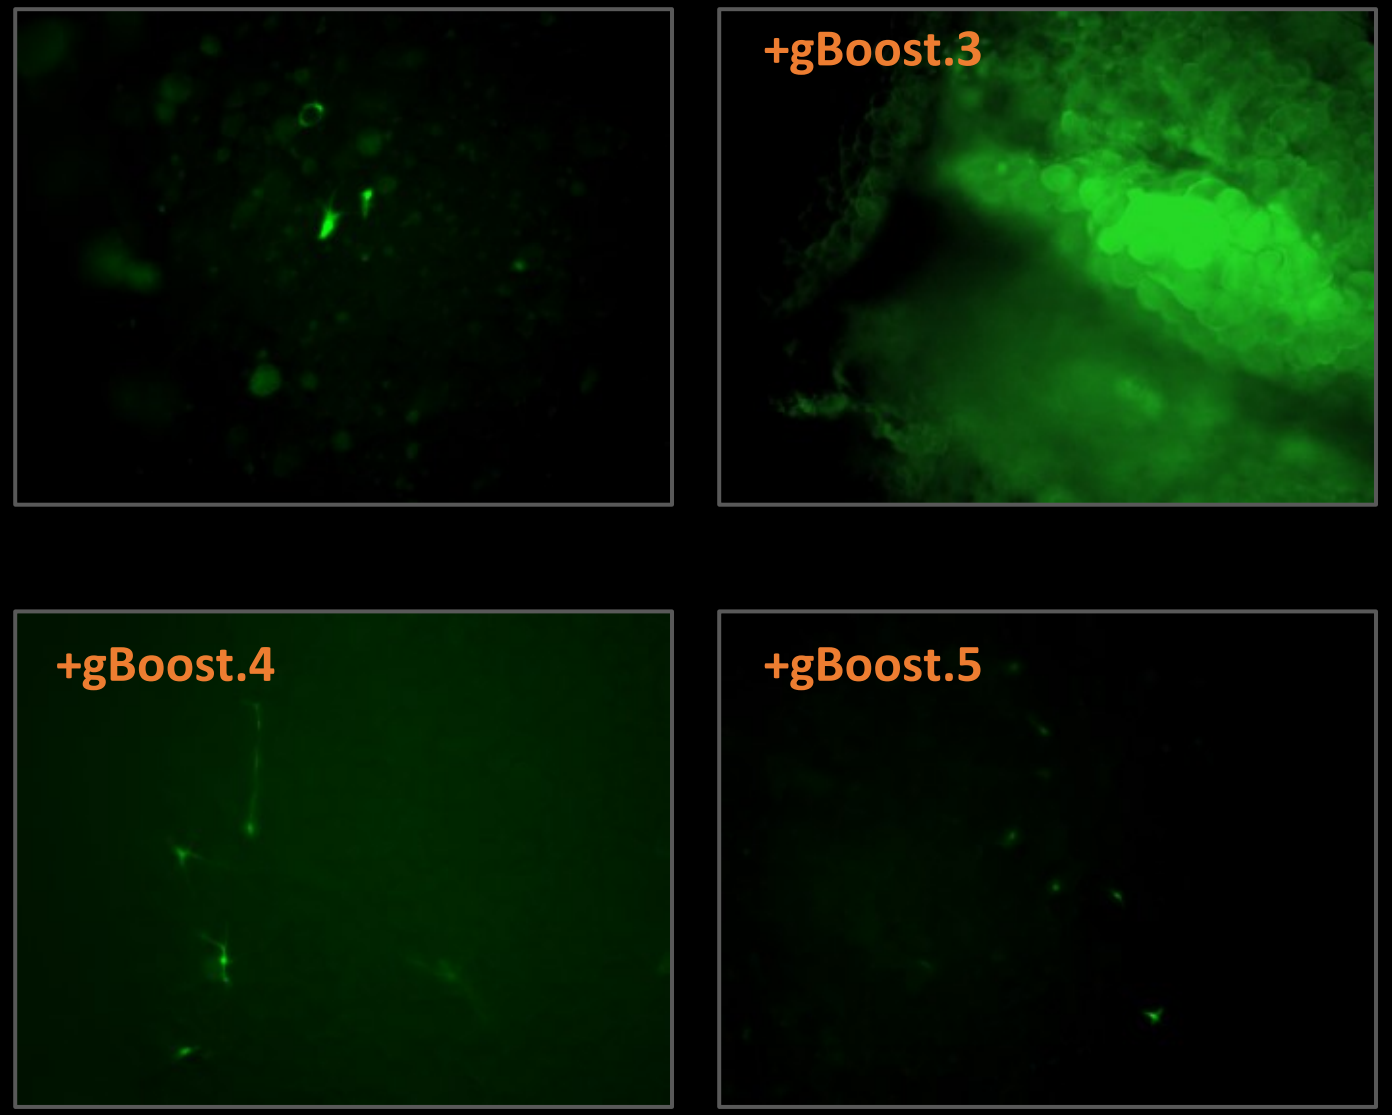
+gBoost.3
+gBoost.4
+gBoost.5

Effects of gBoostTM on non-viral gene expression

Methods: Human subcutaneous fat tissue was injected with 5uL of pEGFP or mRNA_EGFP (200ug/mL) containing gBoost (or without gBoost as control) followed by electroporation with tweezer electrode. The tissue was cultured for the indicated period, and subjected to microscopy analysis (focus x4)

pEGFP, 10d after gene transfer

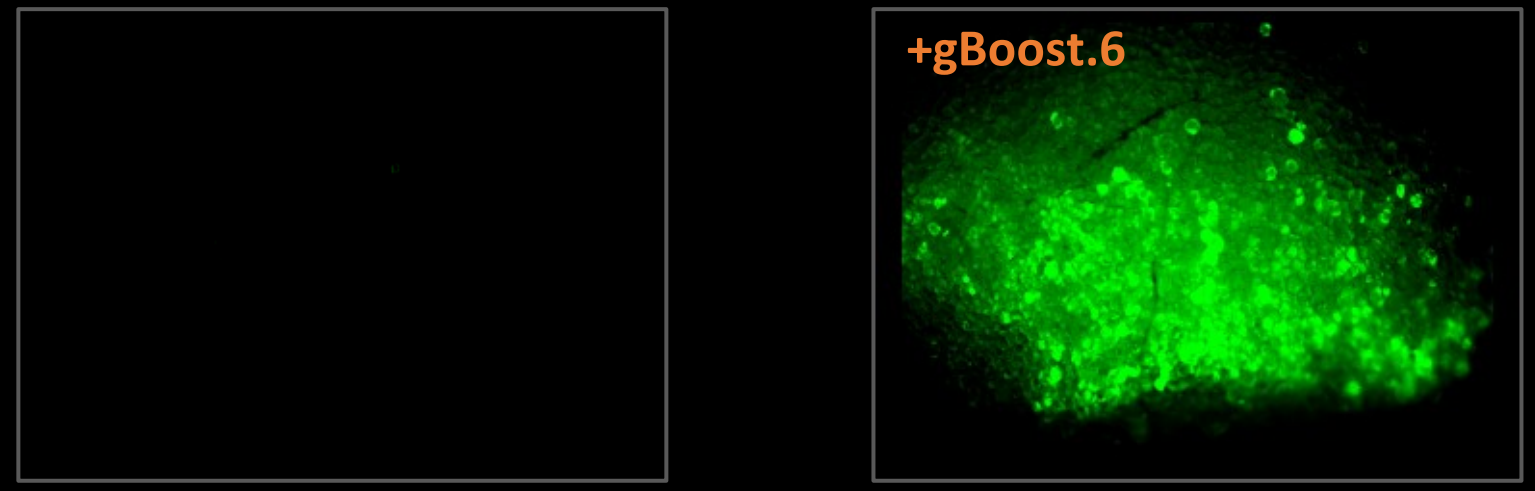
+gBoost.6

mRNA_EGFP, 2d after gene transfer

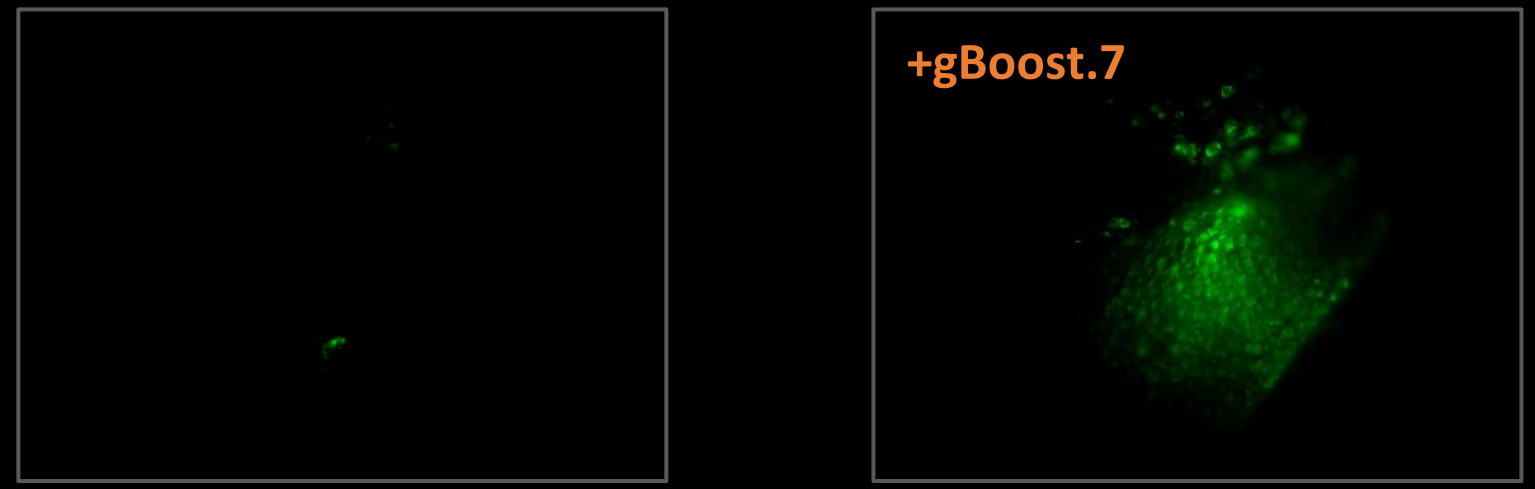
+gBoost.7

gBoost™, a cell-selective gene expression enhancer

Mak Inoue, PhD, Infirmacea Inc. (Chiba, Japan)

Mechanism: inhibition of cellular protective response

Methods: pDNA transfected or AAV5 transducted human fat tissue was cultured for an indicated period. Protective response activity of the tissue was measured (details can be disclosable under NDA).

Time course of protective response

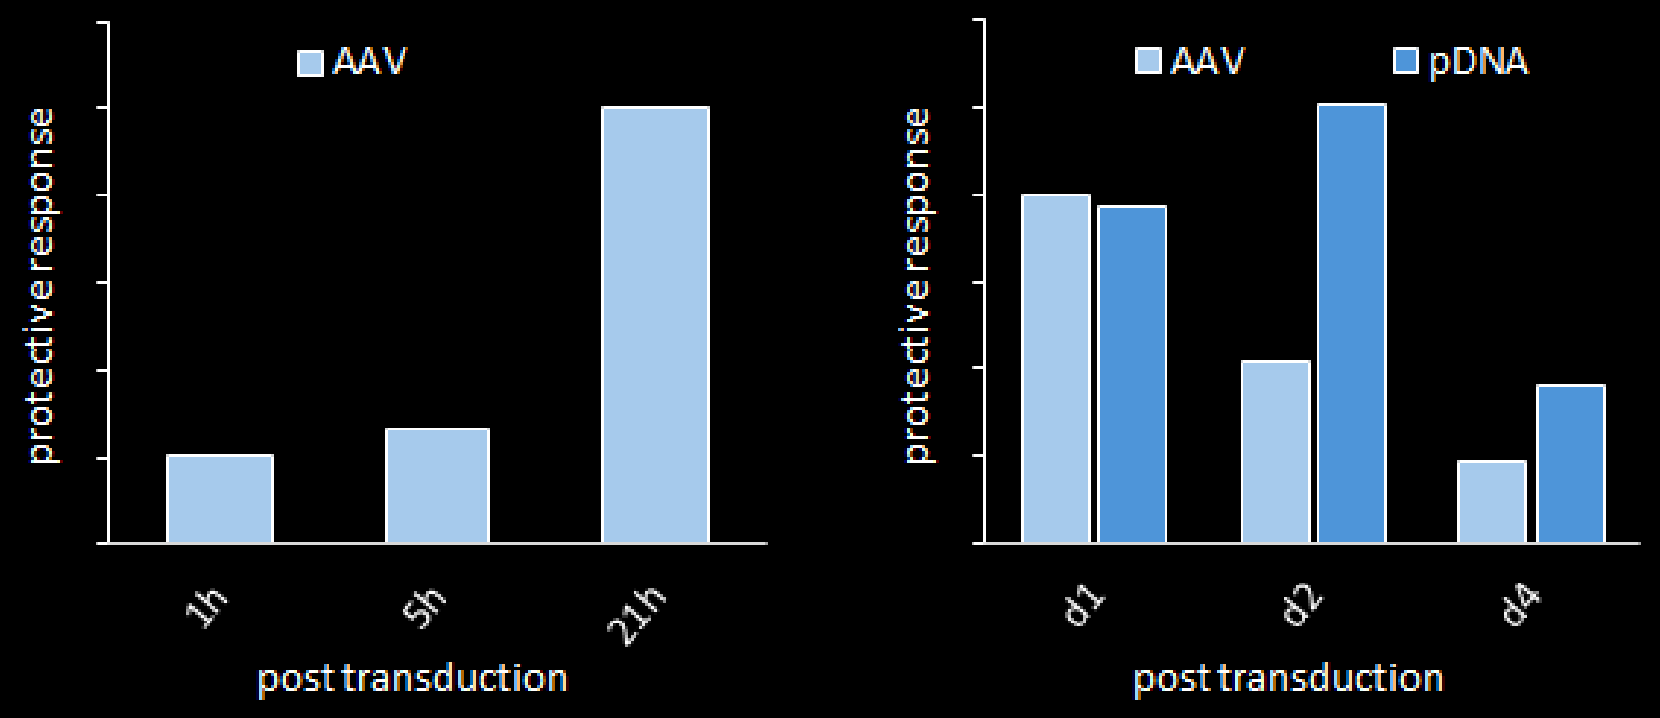
AAV
protective response
1h
5h
21h
post transduction
AAV
pDNA
protective response
d1
d2
d4
post transduction

Findings: Despite lack of gene expression without gBoost, cells response to the transferred genetic materials especially at early phase of transfection/transduction. Effective gBoost (gBoost.3 for AAV or gBoost.6 for pDNA) minimizes the response while ineffective one (gBoost.3 for pDNA) is less efficient.

Effect of gBoost

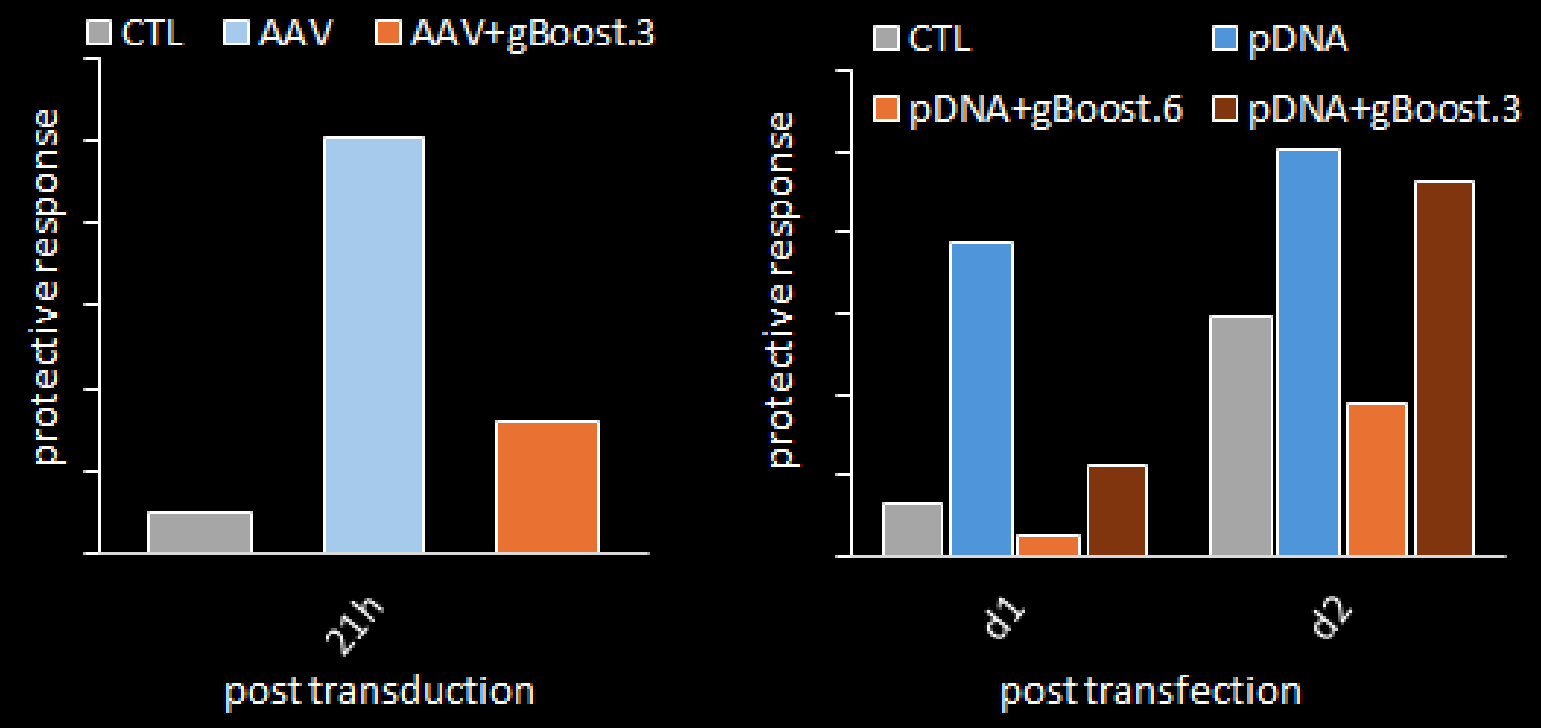
CTL
AAV
AAV+gBoost.3
protective response
21h
post transduction
CTL
pDNA
pDNA+gBoost.6
pDNA+gBoost.3
protective response
d1
d2
post transfection

pDNA-driven protein expression (preliminary)

Methods : Human fat tissue was injected with 5uL of pFGF21 (200ug/mL) containing gBoost (without gBoost as control) followed by electroporation with tweezer electrode. The tissue was cultured for 1 week. Culture medium was exchanged, and after 4 h culture, the medium was collected and subjected to ELISA analysis. The preliminary experiment was carried out as a part of feasibility study for fat-directed DNA therapy as “a safe and affordable” *in vivo* gene therapy.

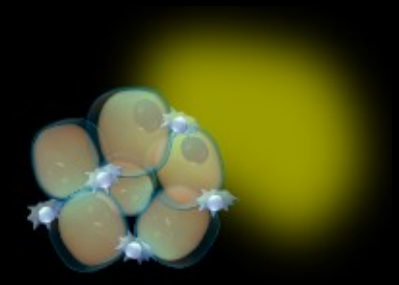

	pg/mL
no transfection	40.7
pFGF21	50.0
pFGF21 + gBoost.7	570.9

In vivo pDNA expression (preliminary)

Methods : mouse was sc injected (targeting inguinal fat) with 10 x3uL of pEGFP (200ug/mL) containing gBoost followed by electroporation with tweezer electrode. After 1 week, mouse was sacrificed, inguinal fat was dissected, and subjected to microscopy analysis (focus x4)

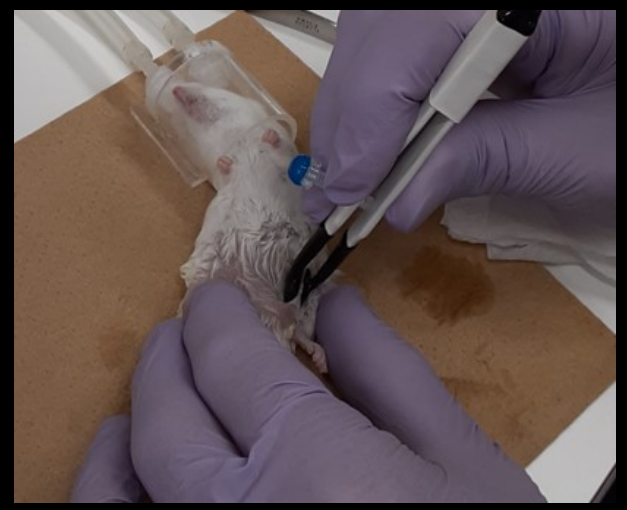

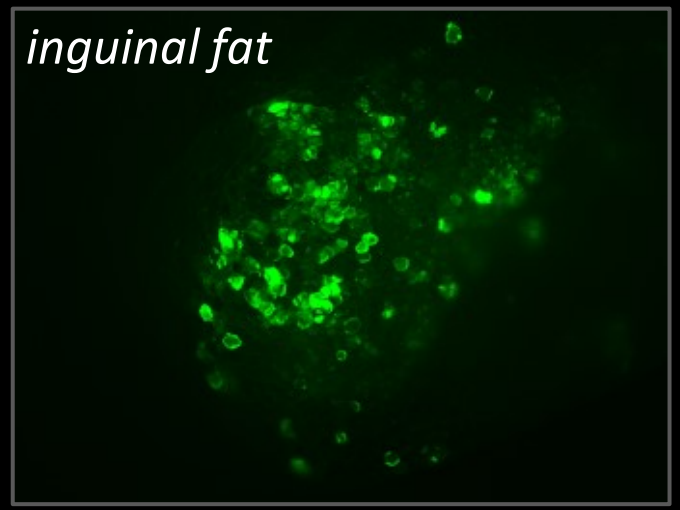
inguinal fat

Conclusion

gBoost™ enhances gene expression either non-viral (pDNA, mRNA) or viral (AAV) in a cell selective manner by minimizing exogenous material-triggered cellular protective response especially at early phase of transfection/transduction. gBoost™ is a combination of drugs, that can be optimized for delivery agent (AAV serotype etc.) and target cells.

Get in touch, follow us

Please feel free to contact us for gBoost trial.

Mak Inoue
CEO at Infirmacea Inc.